CD14 inhibition mitigates myocardial damage and dysfunction following myocardial infarction

A Brown[1], H Kiriazis[1,2], J Noonan[1,2,3], I Hsu[1], G Farrugia[1], H Fang[1], T Yee Tai[1], S Jansen[1], N Carvajal[3], C Krstevski[1], W Shihata[1], YK Tham[1,2,3], C Cohen[3], A Vais[3], A Parslow[1], C Johnson[4], M Imiyage Dona[1], B Wang[1,2,3], Abbate A[5,6], Lavine, K[7,8,9], M Appleby[10], D Crowe[10], B Ziegelaar[10], Redlich G[10], D Greening[1,2,3], A Pinto[1,3,4], D Kaye[1,2,3,11], **D Donner[1,2,3*]**

Corresponding Author: E daniel.donner@baker.edu.au Ph +61(3)85321177 Tw @drdandonner

International Society for Heart Research XXIV World Congress

1 Baker Heart & Diabetes Institute, Melbourne, Australia. 2 University of Melbourne, Melbourne, Australia. 3 Monash University, Melbourne, Australia. 4 La Trobe University, Melbourne, Australia. 5 VCU Pauley Heart Center, Virginia Commonwealth University, Richmond, VA, USA. 6 Berne Cardiovascular Research Centre, University of Virginia, USA. 7 Center for Cardiovascular Research, Department of Medicine, Washington University School of Medicine, St. Louis, MO, USA. 8 Department of Pathology & Immunology, Washington University School of Medicine, St. Louis, MO, USA. 9 Department of Developmental Biology, Washington University School of Medicine, St. Louis, MO, USA. 10 Implicit Bioscience, Seattle, WA, USA. 11 Department of Cardiology, Alfred Hospital, Melbourne, Australia.

Background

- MIR injury causes focal tissue scarring and initiates an excessive immune response leading to scar expansion, adverse remodelling and LV dysfunction, predisposing patients with poor LV function at admission (EF<40) to debilitating heart failure with reduced ejection fraction (HFrEF)[1]
- CD14 is a master regulator of the macrophage response to damage associated molecular patterns (DAMPs) released with MIR
- Pro-inflammatory CD14^{++} macrophages are instrumental to the propagation of adverse inflammatory signals and secondary damage[1]
- Atibuclimab, a novel anti-CD14 monoclonal antibody, has been developed as a safe and selective inhibitor of CD14 without immune suppression

Aims

To assess safety, immunological modulation and cardioprotection with CD14 inhibition given at reperfusion in rigorous murine studies of MIR injury

Hypothesis

Anti-CD14 treatment given at time of reperfusion protects against progressive left ventricular damage and dysfunction, is associated with modulation of macrophages, independently of acute damage

Project Overview

Animal ethics All experiments were approved by the ARA Animal Ethics Committee (E/7610/2019/B).

Study design, models and methods Adult male C57Bl6 mice underwent MIR surgery to occlude the left anterior descending coronary artery for 60mins prior to reperfusion (Fig 1). Mice were randomised and coded, receiving IV saline (SAL, n=50), anti-CD14 antibody (n=50) or isotype control (ISO, n=50) at reperfusion and again at 24h. Assessments included infarct size (by Evan's blue/TTC), plasma troponin, bulk tissue RNA seq, flow cytometry, single cell RNA seq, histological fibrosis, immunohistochemistry, LV echocardiography (PLAX), and LV pressure-volume catheterisation at 1, 3 or 7 days post-MIR.

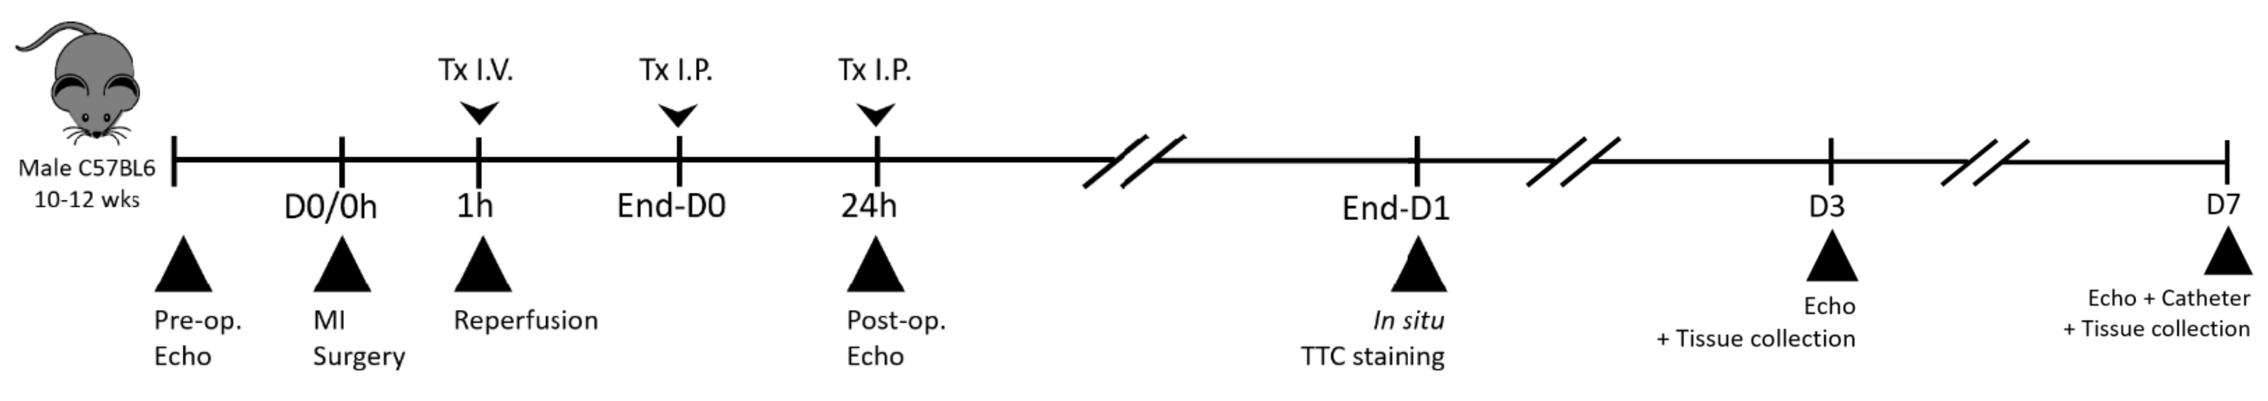
Male C57BL6 10-12 wks
Tx I.V.
Tx I.P.
Tx I.P.
D0/0h
1h
End-D0
24h
End-D1
D3
D7
Pre-op. Echo
MI Surgery
Reperfusion
Post-op. Echo
In situ TTC staining
Echo + Tissue collection
Echo + Catheter + Tissue collection

Fig 1. Three-phase preclinical trial design Hypothesis-driven studies were performed in series and parallel with independent cohorts for End-D1, D3 and D7 endpoints.

Statistics One-way ANOVA and Tukey (parametric) or Kruskal-Wallis (non-parametric) post-hoc tests were applied while blinded. All graphed data presented as mean±SD, in-text as mean±SE.

Preclinical Trial Rigour & Reproducibility

All mice were **randomised** and each study **independently blinded** (treatments coded to A, B, C) throughout all *in vivo* and post-mortem procedures, data acquisition, data analysis and reporting. **Exclusion criteria** were predetermined to be model insufficiency (lack of ST-segment elevation during surgery [Fig 2] or area-at-risk at 24h <35% [Fig 3]) or technical insufficiency of endpoint (e.g. ruptured vessel during catheterisation).

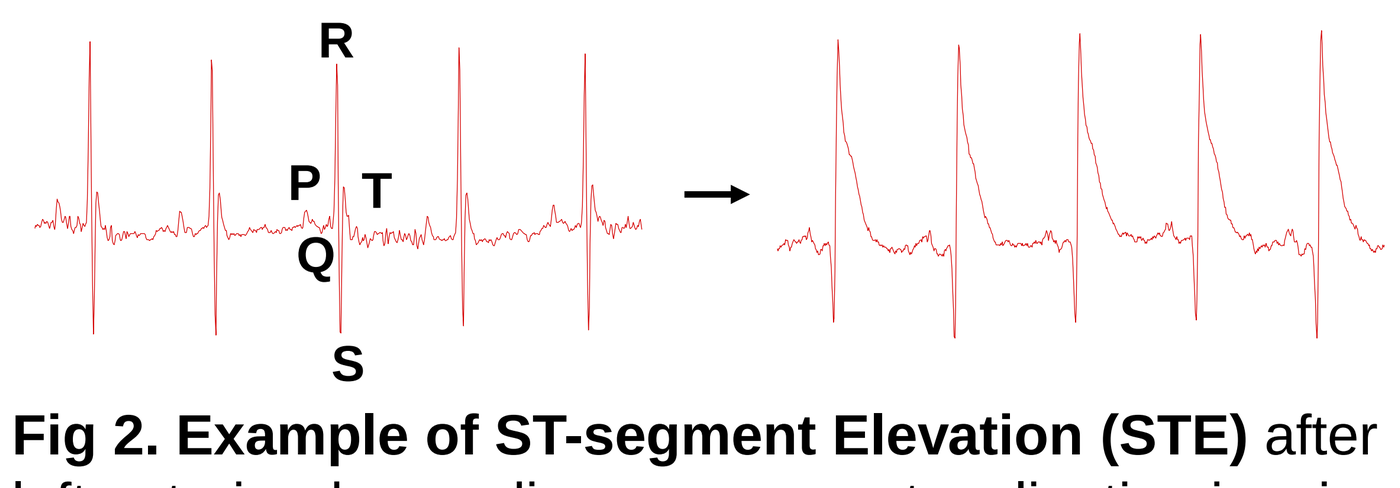
R
P
T
Q
S

Fig 2. Example of ST-segment Elevation (STE) after left anterior descending coronary artery ligation in mice

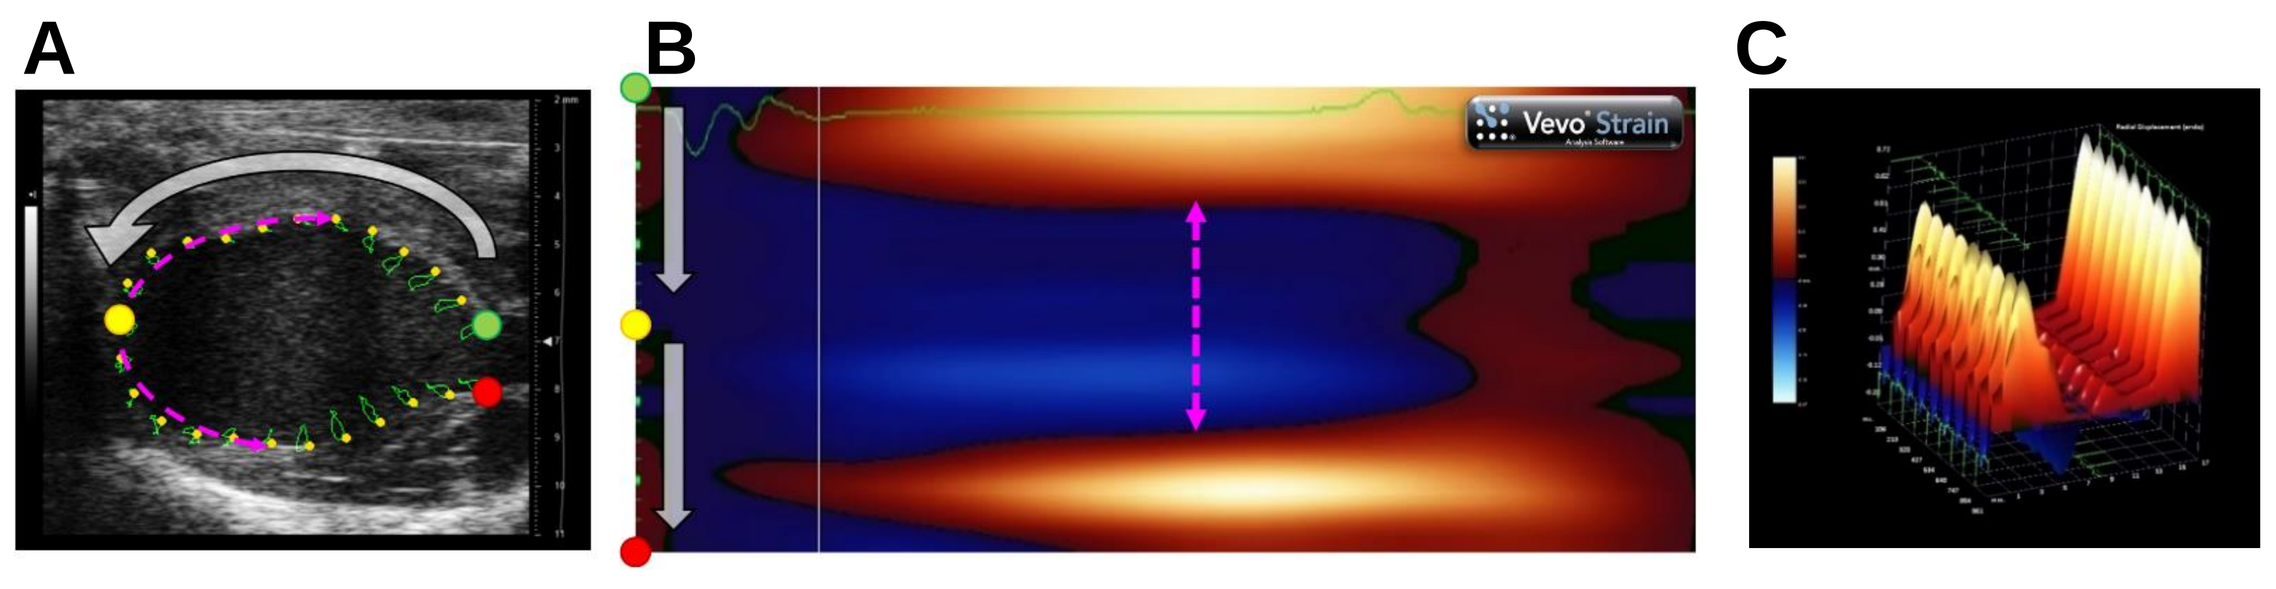
A
B
C
Vevo Strain

Fig 3. Assessment of area-at-risk (AAR) by echocardiographic strain at 24h post-MIR surgery A) Example tracing of the endocardium, B) relative displacement map with dotted pink line denoting ischemic zone,[2] C) 3-dimensional representation of displacement between cardiac cycles (parallel waveforms)

The **primary endpoint** for each separately delivered study was pre-specified as infarct size (end-D1 study), immune cell phenotype by single cell RNA seq (D3), and LV ejection fraction (EF%) by ultra-high frequency echocardiography (D7), for relevant power calculations.

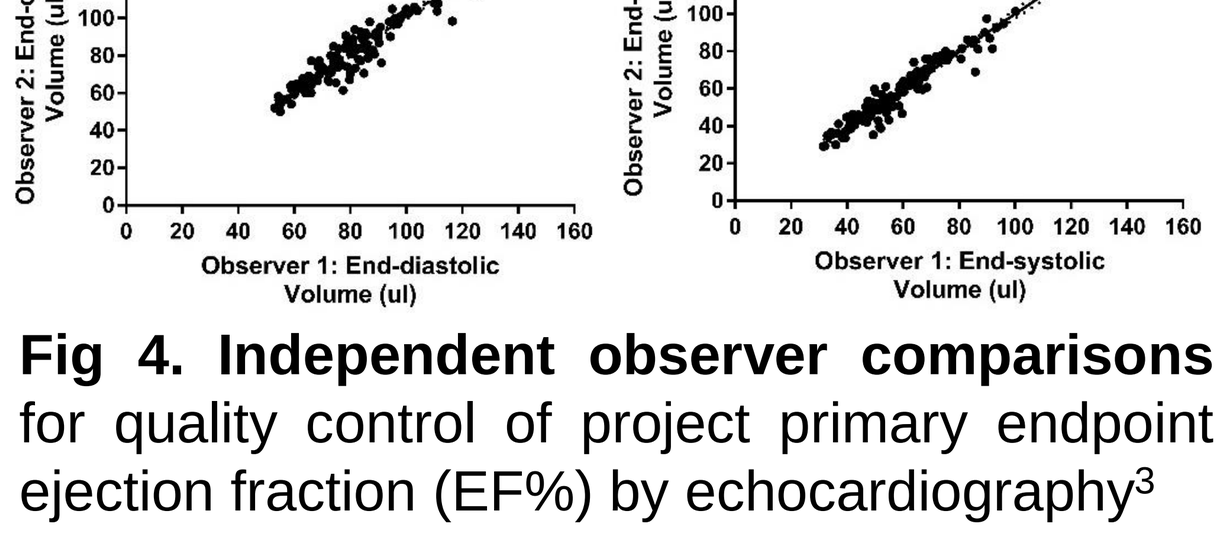
slope = 1.0 ± 0.0
r² = 0.9
Observer 2: End-diastolic Volume (ul)
Observer 1: End-diastolic Volume (ul)
slope = 1.0 ± 0.0
r² = 0.9
Observer 2: End-systolic Volume (ul)
Observer 1: End-systolic Volume (ul)

Fig 4. Independent observer comparisons for quality control of project primary endpoint ejection fraction (EF%) by echocardiography[3]

References

1. Frantz et al. (2022) Left ventricular remodelling post-myocardial infarction: pathophysiology, imaging, and novel therapies. *Eur Heart J.* 00, 1-16.
2. **Donner** et al. (2020) A novel non-invasive method of measuring myocardial infarct size in the mouse using cardiac ultrasound. *J Mol Cell Cardiol*. 140, 6-7
3. **Donner** et al. (2018) Improving the quality of preclinical research echocardiography: observations, training and guidelines for measurement. *Am J Physiol-Heart & Circ Physiol.* 315(1), H58-70.

Findings

*Clinically significant protection against left ventricular dysfunction at **7 days** post-MIR*

Compared to SAL and ISO, at 7d post-MIR surgery (n=3grps x 15/grp), anti-CD14 treatment significantly improved LV EF by >30 % (33±2 vs 25±1 in SAL, 24±2 % in ISO, p<0.001); stroke work by >38 % (1662±69 vs 1103±63 in SAL, 1201±96 mmHg x ul in ISO, p<0.001), -dV/dT by >24 % (-867±67 vs 563±30 in SAL, 700±59 ul / s in ISO, p<0.01) and peak power by >70 % (14.4±2.8 vs 6.7±0.7 in SAL, 8.4±1.1 mmHg x ml / s in ISO, p<0.05), see Fig 5A-D.

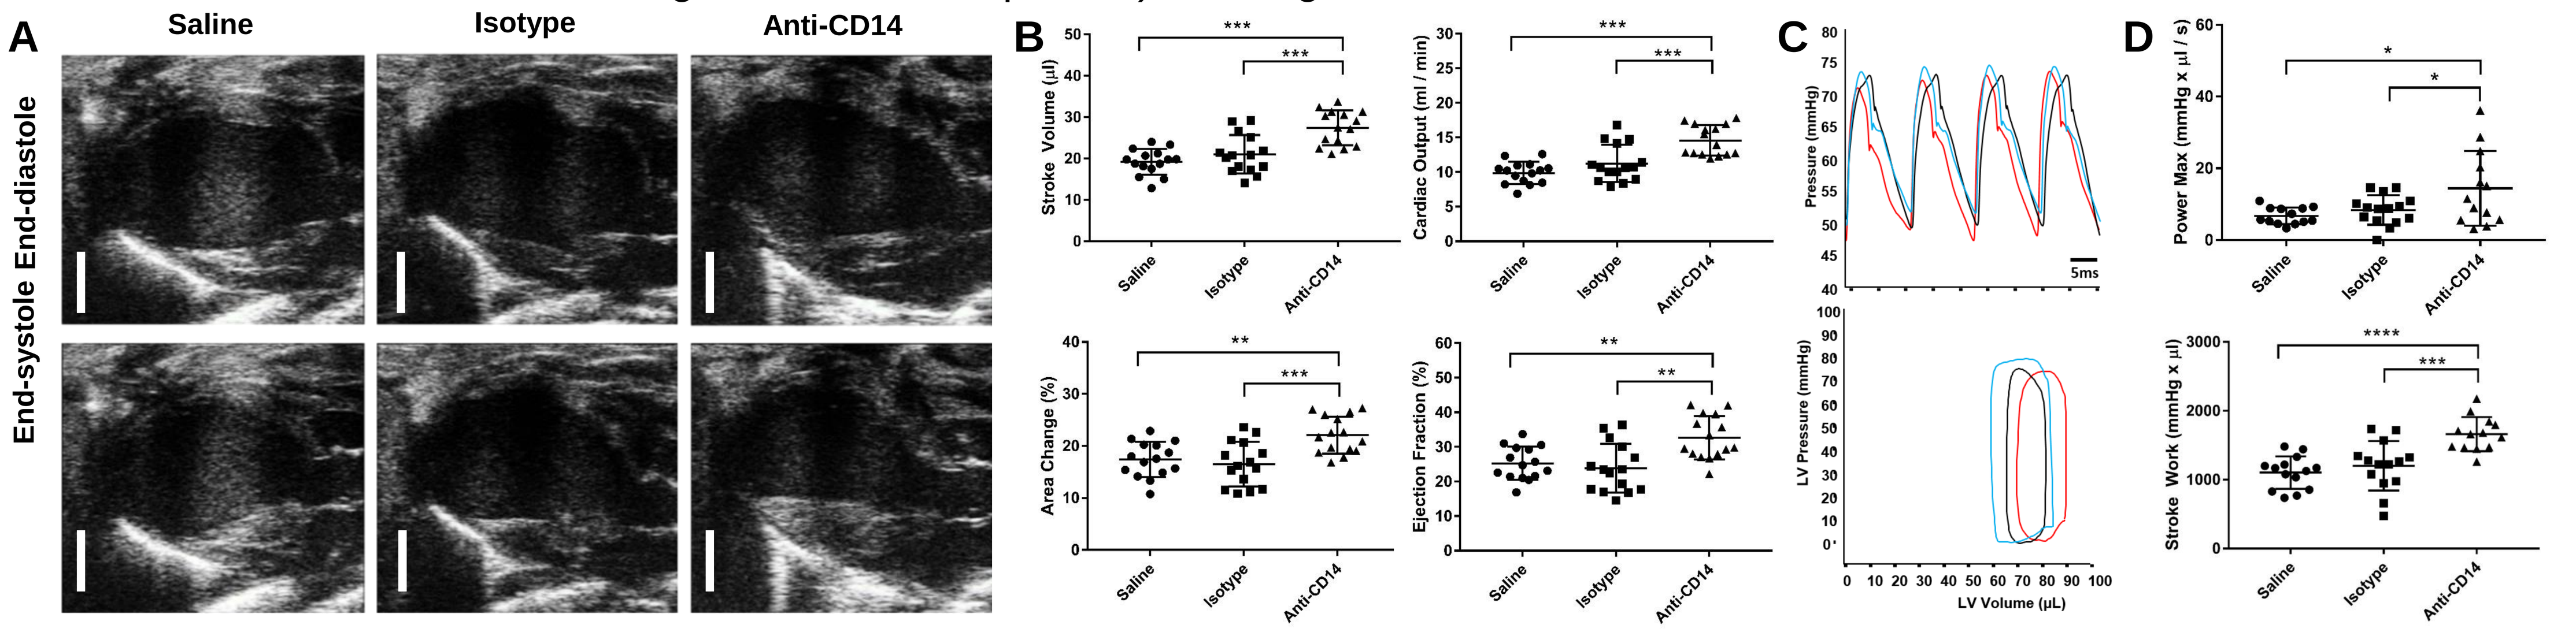
A
Saline
Isotype
Anti-CD14
End-systole End-diastole
B
Stroke Volume (µl)
Cardiac Output (ml / min)
Area Change (%)
Ejection Fraction (%)
C
Pressure (mmHg)
5ms
LV Pressure (mmHg)
LV Volume (µL)
D
Power Max (mmHg x µl / s)
Stroke Work (mmHg x µl)

Fig 5. LV function 7 days post-MIR A) Representative echocardiographic images at end-diastole and -systole, B) corresponding echocardiographic parameters, C) representative arterial blood pressures (upper) and pressure-volume loops (lower), and D) corresponding haemodynamic parameters. Scale bar 2mm.

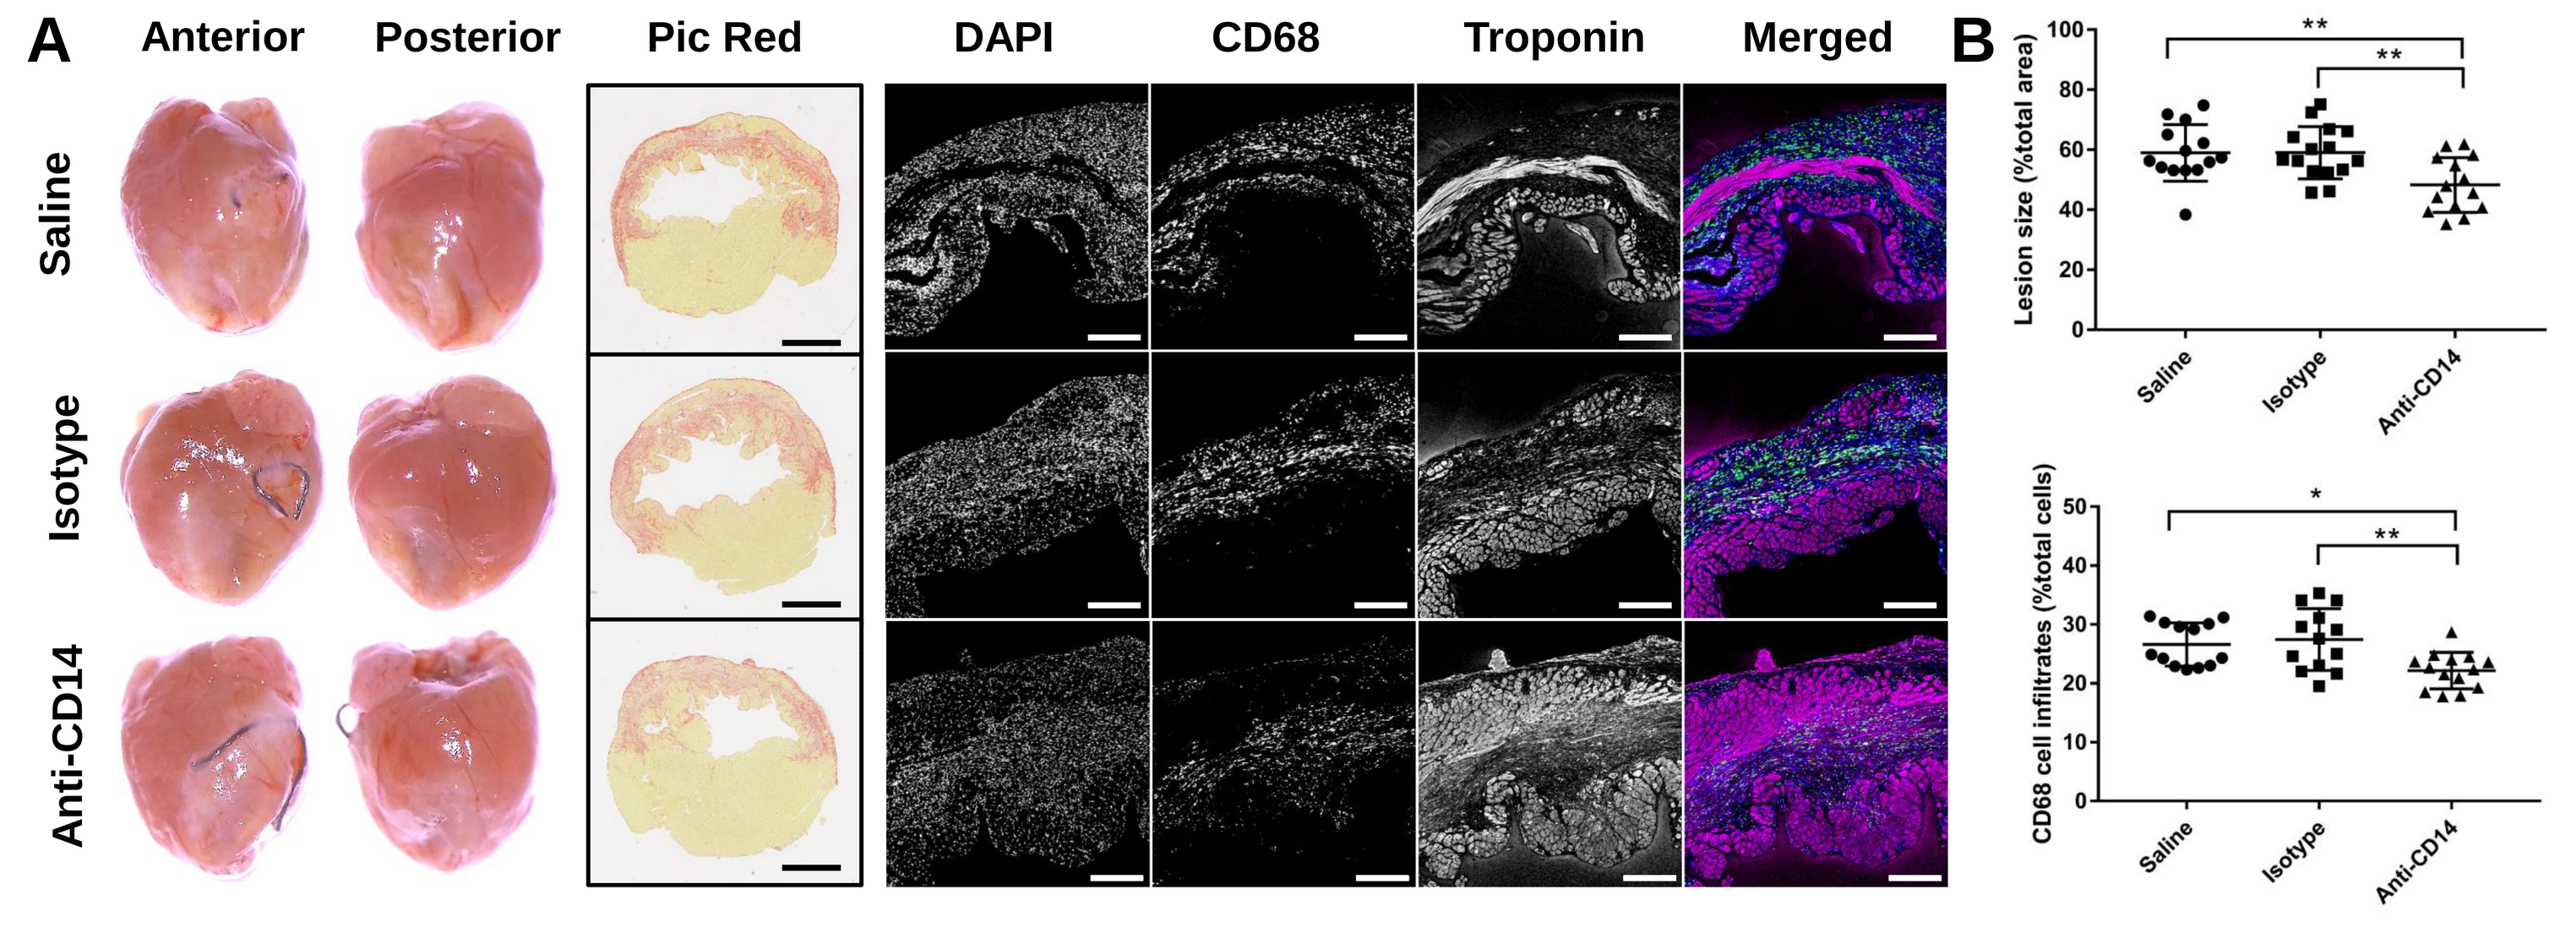
A
Anterior
Posterior
Pic Red
DAPI
CD68
Troponin
Merged
Saline
Isotype
Anti-CD14
B
Lesion size (%total area)
CD68 cell infiltrates (%total cells)

Fig 6. Post-mortem tissue imaging A) From left – right: whole heart photgraphs, brightfield imaging of mid-ventricular section stained with picrosirius red, and immunofluorescent imaging of mid-ventricular section with channels for DAPI (blue), CD68 (green), troponin-I (pink) and merged, B) corresponding histological parameters lesion size (by picrosirius red staining) and CD68+ cell infiltrate abundance (immunohistochemistry). Scale bar 200µm.

Anti-CD14 treatment significantly reduced scar size (48.2±2.5 vs 58.9±2.5 in SAL, 59.0±2.2 % total area in ISO, p<0.01) and CD68+ infiltrates (22.2±0.8 vs 26.6±1.0 in SAL, 27.4±1.5 % total cells in ISO, p<0.01). No differences were observed for heart, lung, kidney, or spleen weights at 7 days post-MIR. No rupture was observed in any mice in these studies, indicating consistently of reperfusion in model (equivalent to re-establishing vessel patency clinically with Percutaneous Coronary Intervention; PCI), as well as tx safety.

*Independent of acute infarct size or injury at **1 day** post-MIR*

No differences were observed in area-at-risk or infarct size (measured by Evan's blue/TTC dual staining method, Fig 7A), or circulating cardiac troponin-I (cTnI, by commercial ELISA kit, Fig 7B) at 1 day post-MIR surgery.

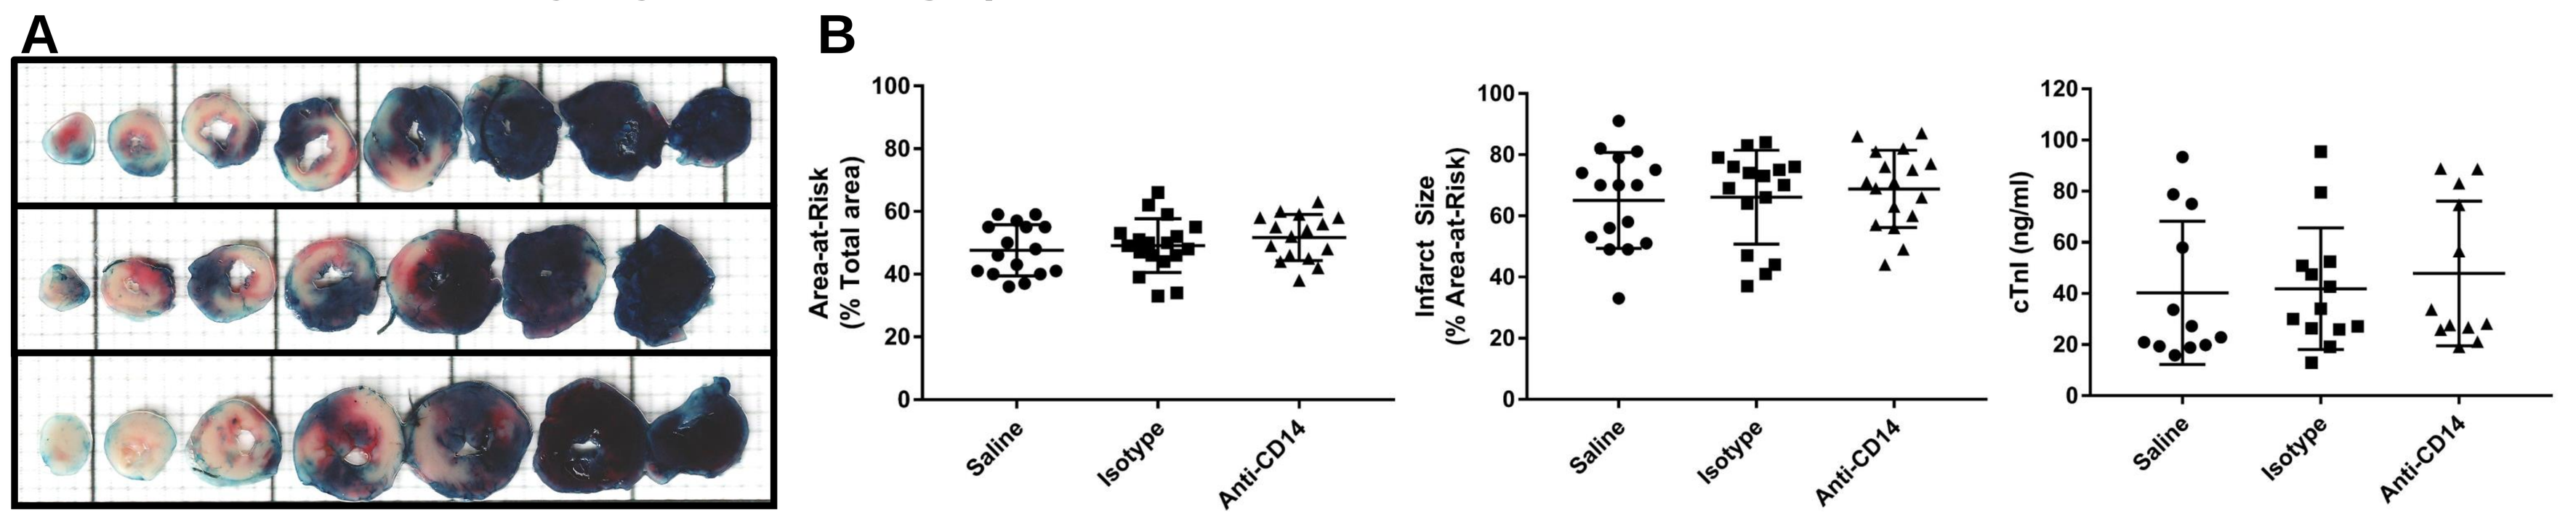
A
B
Area-at-Risk (% Total area)
Infarct Size (% Area-at-Risk)
cTnI (ng/ml)
Saline
Isotype
Anti-CD14

Fig 7. Acute cardiac injury 1 day post-MIR A) Representative images of Evan's blue and triphenyl tetrazolium chloride (TTC) co-stained heart sections. Non-ischemic myocardium is indicated by the blue-stained, ischemic myocardium (Area-at-Risk; AAR) is indicated by the sum of pink and white/beige, and infarct size (IS) is indicated by white/beige areas B) Corresponding AAR, IS and cTnI comparisons.

*Associated with modulation of macrophage population phenotypes at **3 days** post-MIR*

No differences were observed in area-at-risk, fibrosis, relative abundances of viable leukocyte sub-populations counted by either flow cytometry or immunohistochemistry of CD68+ cells in whole left mid-ventricular sections; or any measure of echocardiographic LV function at 3 days post-MIR (all ANOVA p>0.05), data omitted for space. **However**, principal component analysis of bulk tissue RNA sequencing (RNAseq) data indicated deviations from saline and isotype controls with anti-CD14 treatment, further explored with single cell RNAseq (corrected for false discovery rate, p<0.01), shown below.

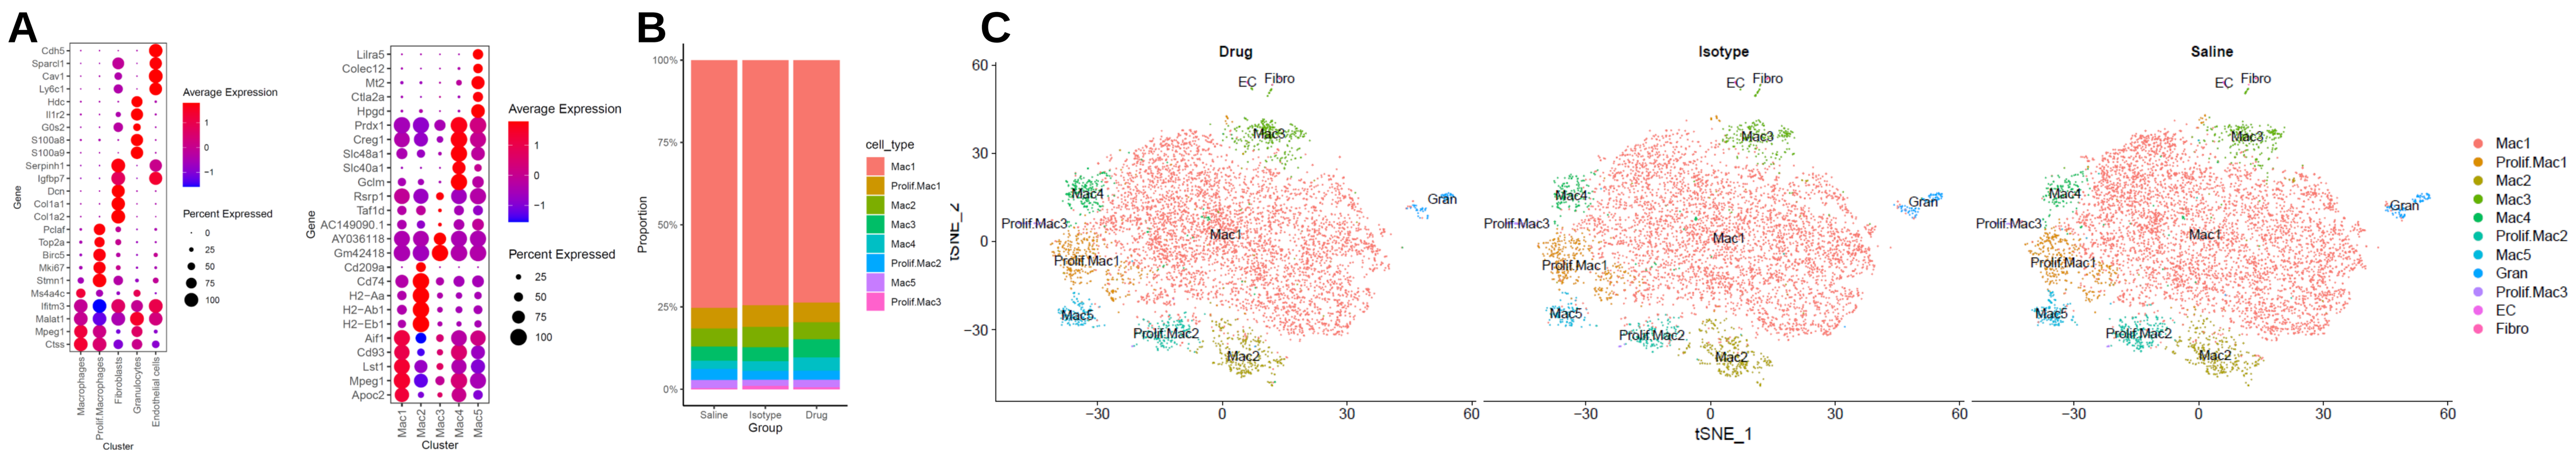
A
Average Expression
Percent Expressed
Cluster
B
Proportion
Group
Saline
Isotype
Drug
cell_type
C
Drug
Isotype
Saline
Mac1
Prolif.Mac1
Mac2
Mac3
Mac4
Prolif.Mac2
Mac5
Gran
Prolif.Mac3
EC
Fibro
tSNE_1
tSNE_2

Fig 8. Single Cell RNA Sequencing at 3 days post-MIR A) Marker gene analysis for cell type (left) and macrophage (right) clustering, B) cell type proportions, C) tSNE cellular atlas plots of sorted macrophage subpopulations.

Significantly differentially expressed (DE) marker genes for each cell subset are shown (Fig 8A). Cell type proportions were similar between groups, with Mac 1 cells representing approximately 75% of total sorted cells in each group (Fig 8B and 8C).

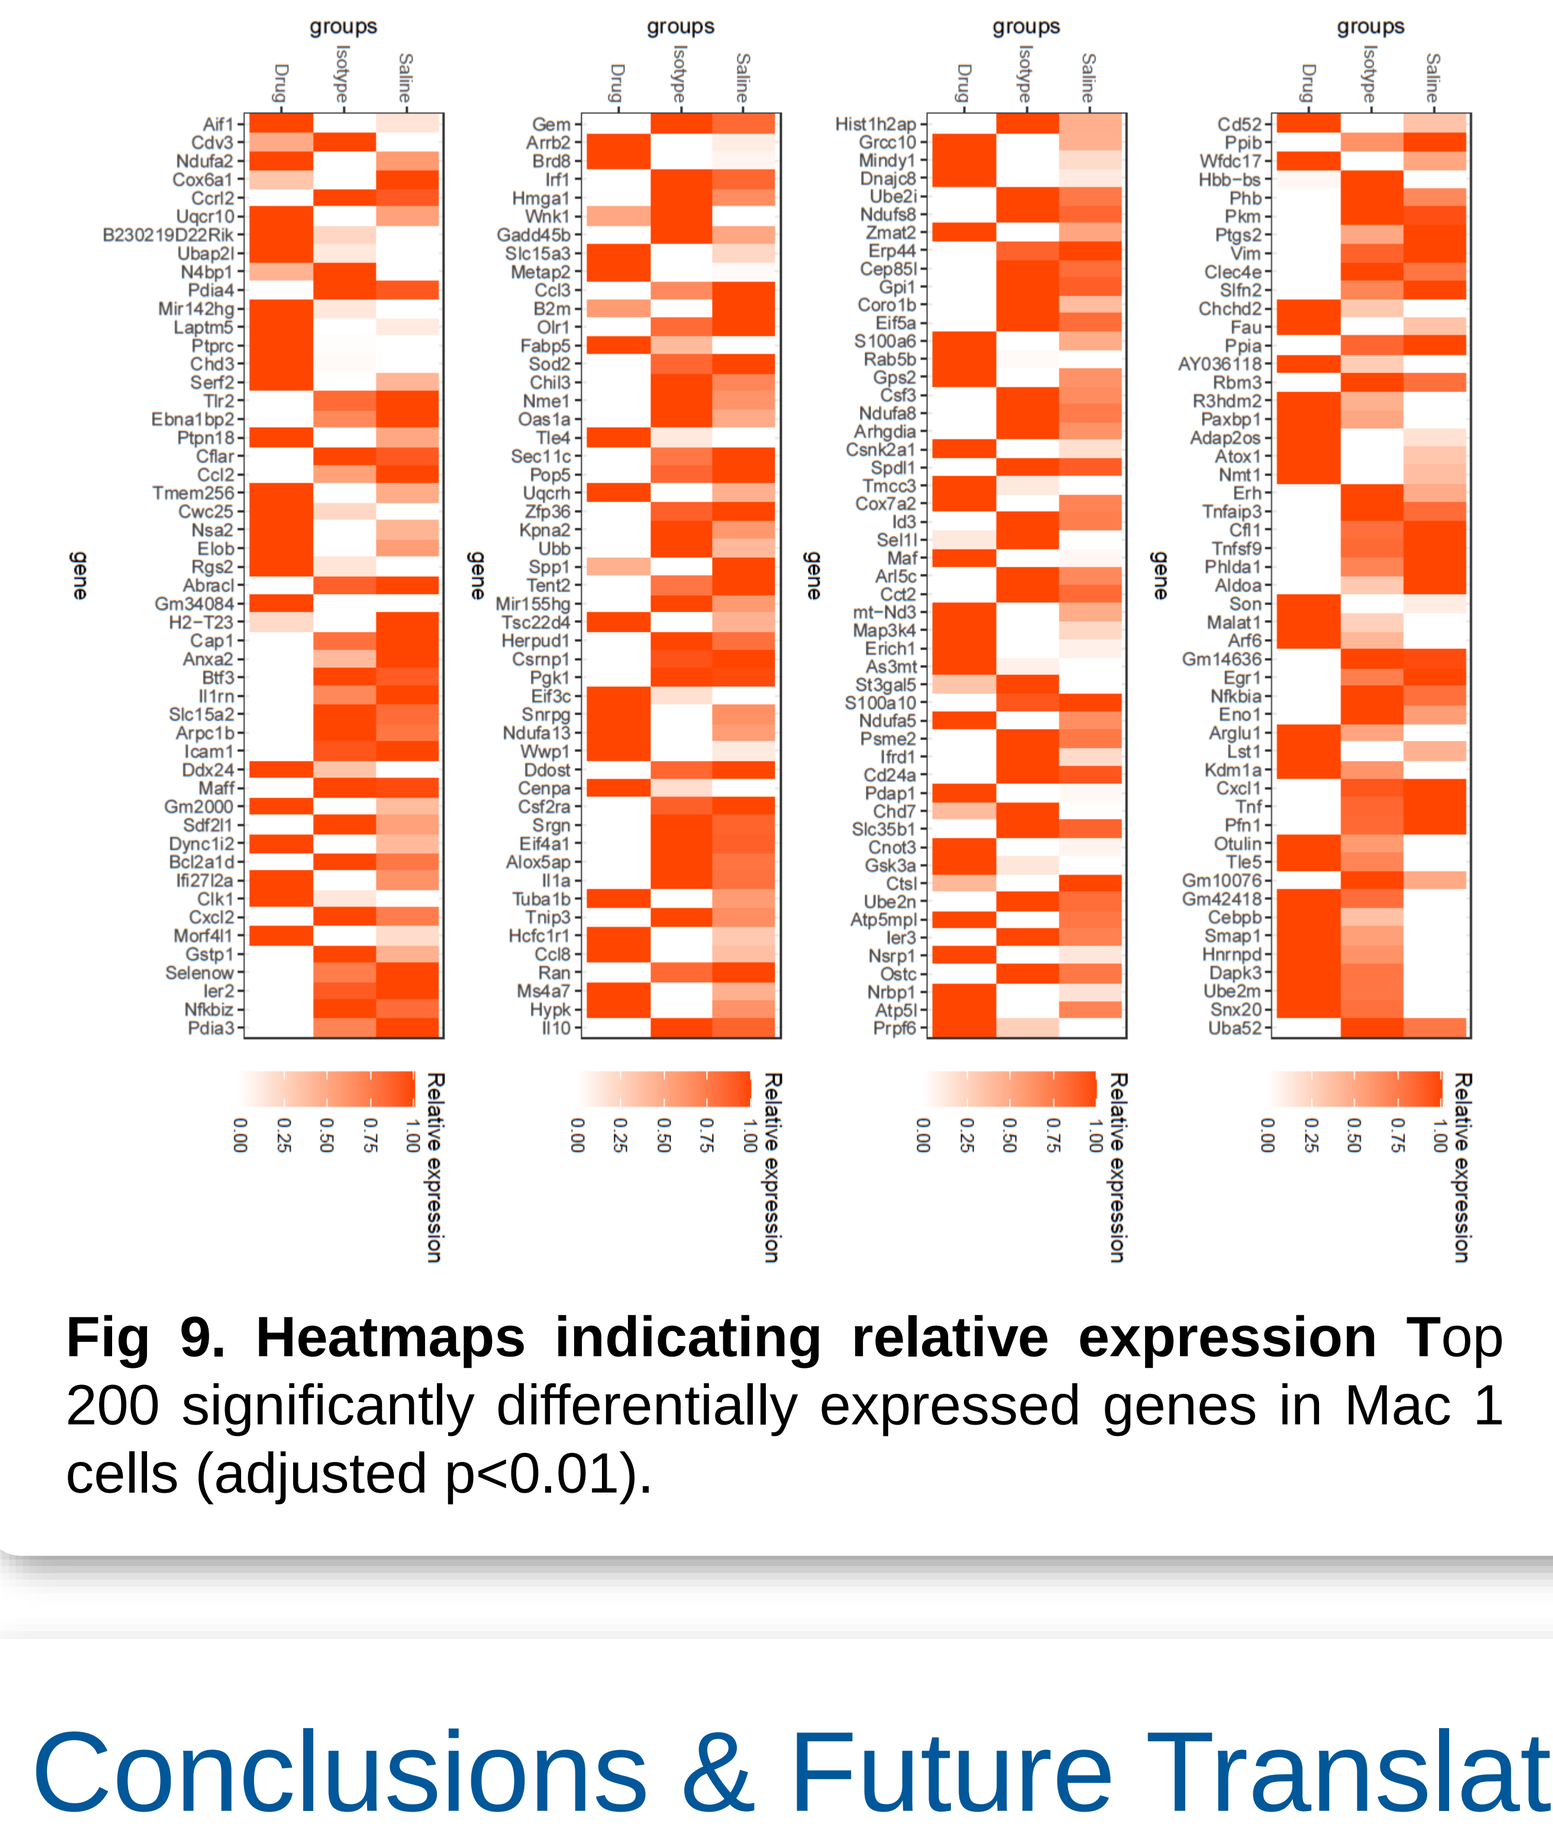
groups
Drug
Isotype
Saline
gene
Relative expression

Fig 9. Heatmaps indicating relative expression Top 200 significantly differentially expressed genes in Mac 1 cells (adjusted p<0.01).

Anti-CD14 treatment resulted in over 1,000 significantly DE genes in macrophage populations. The differences in expression of the top 200 DE genes (ranked by sig.) was predominantly in Mac1 cells (Fig 9). Functional enrichment analysis revealed significant upregulation of genes associated with mRNA processing and chromosome/chromatin modification (omitted for space) and downregulation of large clusters of genes associated with inflammatory responses to damage and other stimuli; immune cell activation, differentiation and migration/chemotaxis; and responses to/production of pro-inflammatory cytokines (Fig 10).

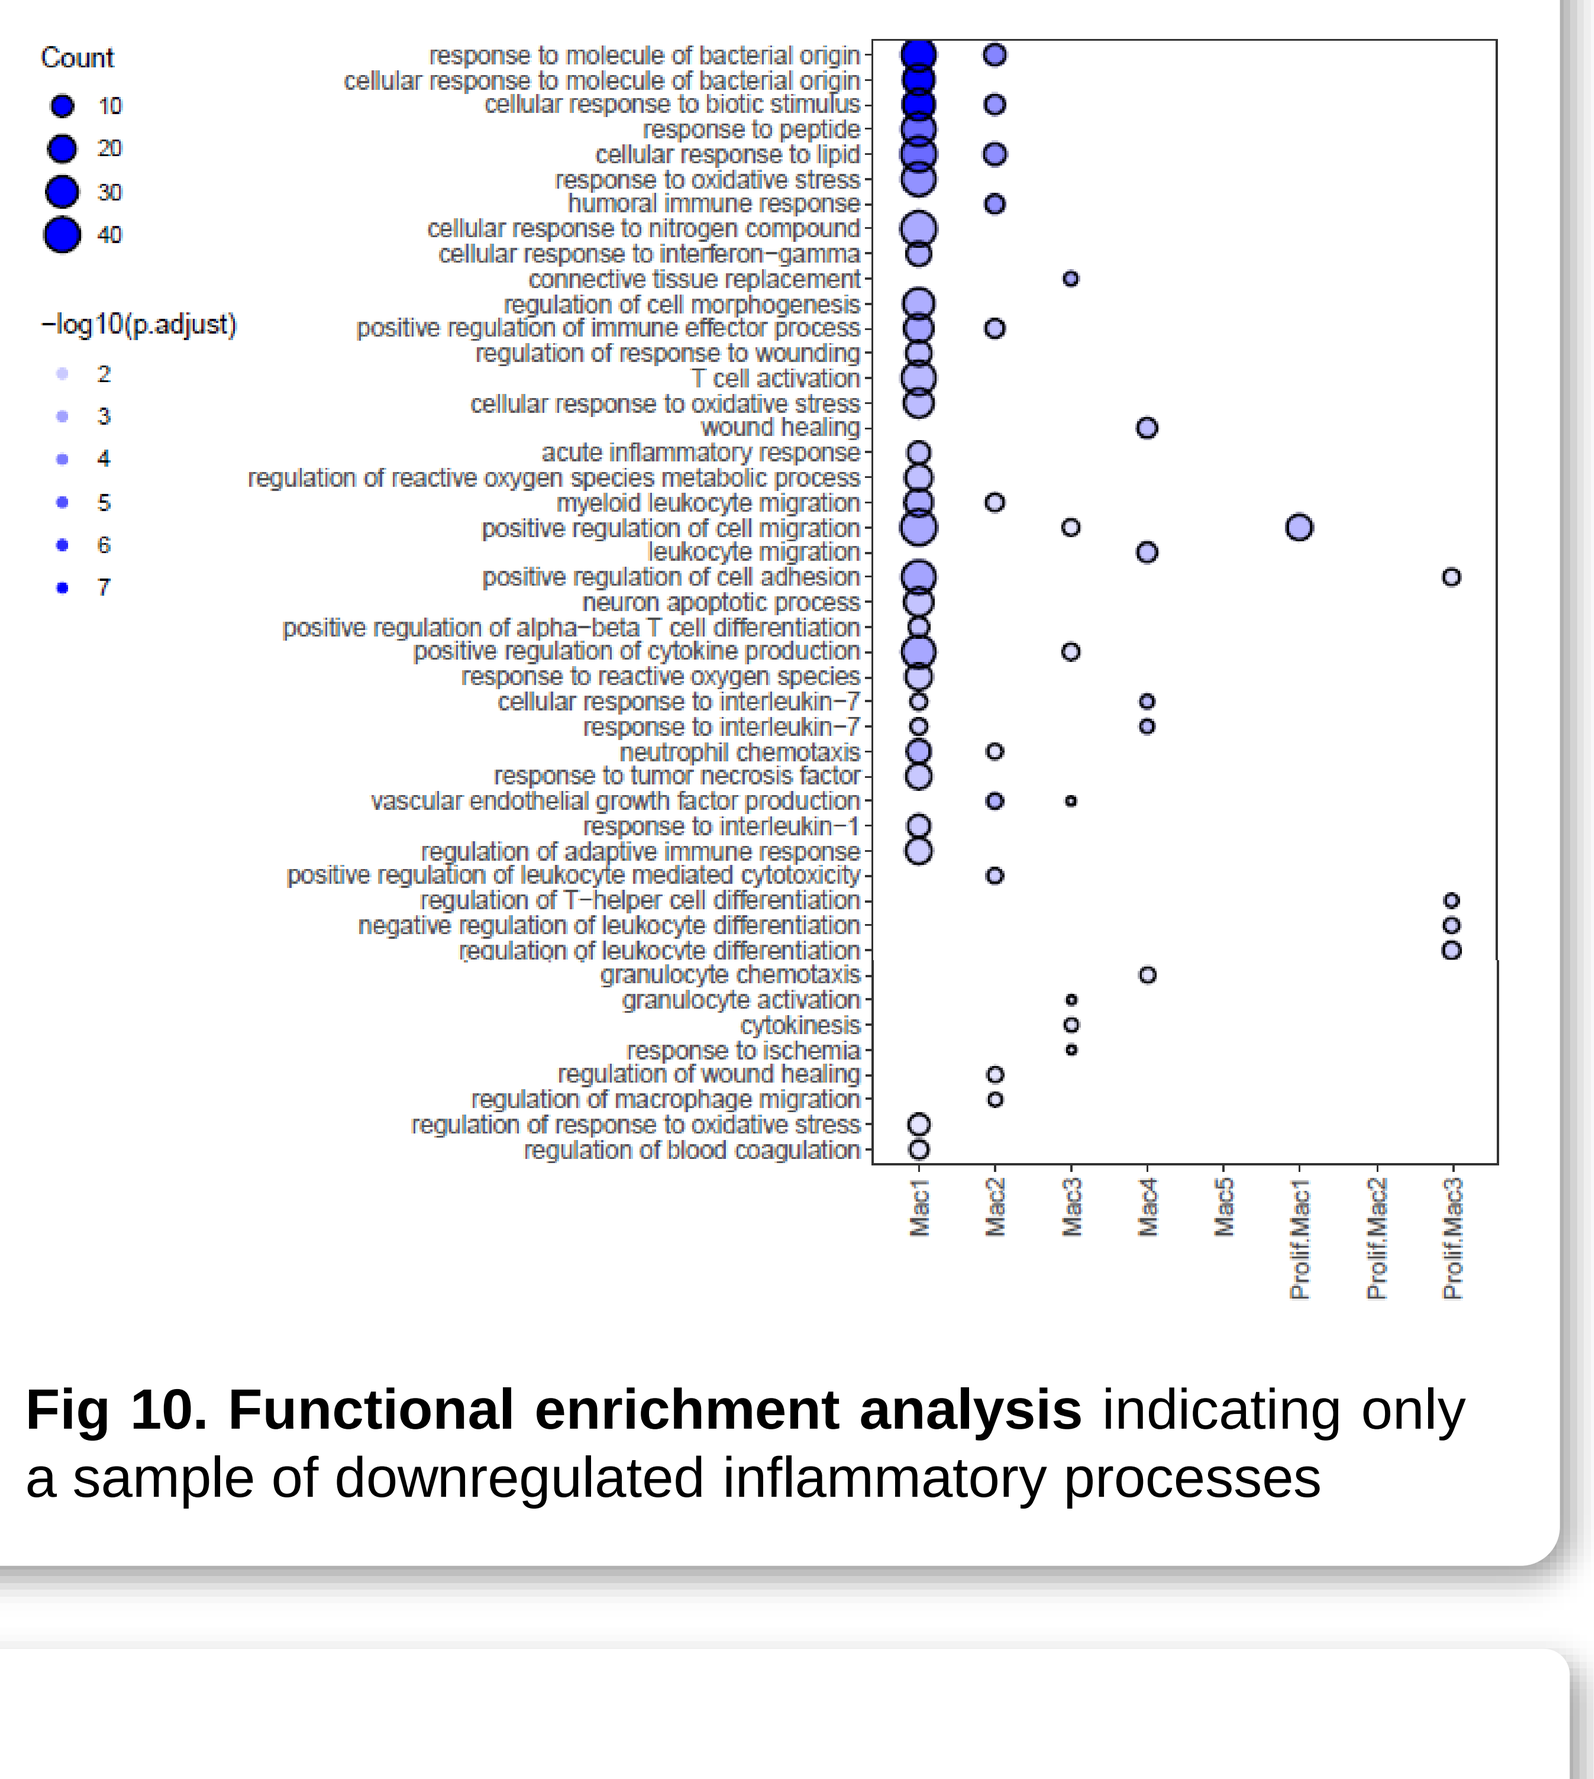
Count
−log10(p.adjust)
Mac1
Mac2
Mac3
Mac4
Mac5
Prolif.Mac1
Prolif.Mac2
Prolif.Mac3

Fig 10. Functional enrichment analysis indicating only a sample of downregulated inflammatory processes

Conclusions & Future Translation

Anti-CD14 treatment given at reperfusion:

- significantly protects against progressive left ventricular injury and dysfunction 7 days post-MIR, independently of acute infarct size
- is associated with modulation of macrophage phenotype at 3 days and subsequent macrophage abundance at 7 days post-MIR

These data support that CD14 antagonism with novel antibody therapies like Atibuclimab may find clinical utility in the protection against immunological drivers of HFrEF in acute MI patients, particularly for those with poor LV function following PCI.

This work formed the basis of successful application for IND and *Study May Proceed* Letter from the FDA for Phase I Clinical Trials